Estimation of equipotent doses of the oral selective glucocorticoid receptor modulator (oSGRM) AZD9567 and prednisolone based on ex vivo TNFα inhibition after LPS stimulation

Joachim Almquist*, Jacob Leander, Waqas Sadiq, Tove Hegelund Myrbäck, Susanne Prothon, Ulf Eriksson

Clinical Pharmacology & Safety Sciences, R&D, AstraZeneca, Gothenburg, Sweden. *Email: joachim.almquist@astrazeneca.com

I-12

Conclusions

- An equipotency relationship between AZD9567 and prednisolone was estimated using PKPD modeling of an ex vivo biomarker of anti-inflammatory effect.
- This relationship is useful for dose selection with respect to efficacy, and provides a common reference system for evaluating safety biomarkers.

Introduction

- AZD9567 is an oral selective glucocorticoid receptor modulator (oSGRM) aimed for the treatment of inflammatory disease [1].
- Designed to deliver similar efficacy to oral steroids such as prednisolone but to differentiate on safety.
- We present a model-based approach to estimate equipotent doses of AZD9567 and prednisolone in terms of equally large inhibition of an ex vivo biomarker for anti-inflammatory effect (TNFα).
- The equipotency relationship makes it possible to translate doses of one compound into the other based on their anti-inflammatory effect.

Objectives

- To estimate an equipotent dose relationship between AZD9567 and prednisolone with respect to ex vivo TNFα inhibition.
- Key intermediate steps were the development of population pharmacokinetic (PK) models for each compound and a TNFα population pharmacodynamic (PD) model for both compounds.

Equipotent doses of AZD9567 and prednisolone estimated from PKPD modeling

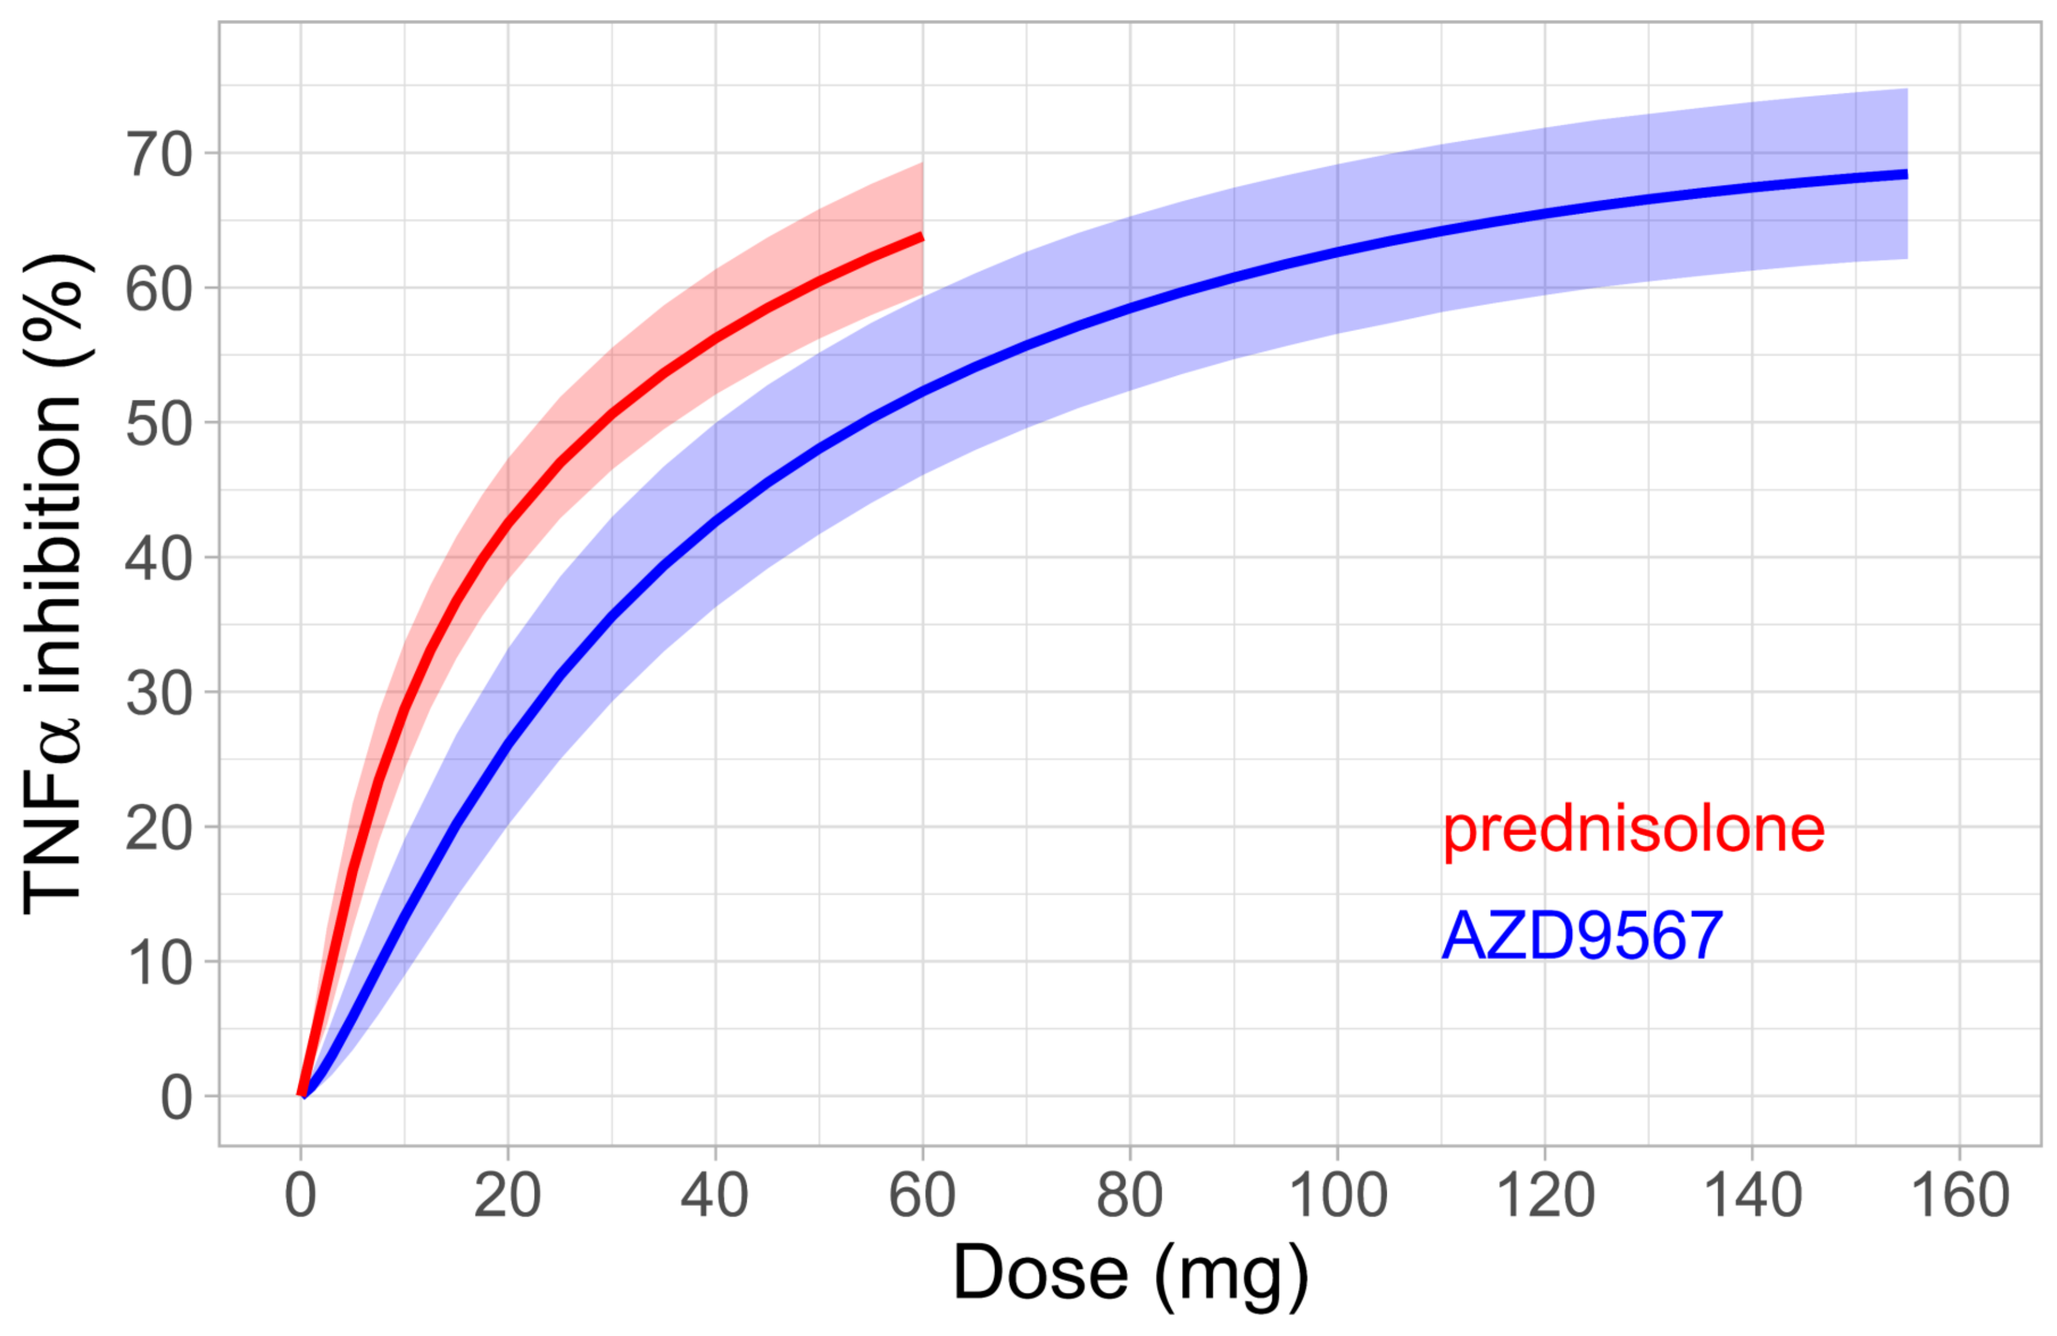
TNFα inhibition (%)
Dose (mg)
prednisolone
AZD9567

Estimated dose-response of AZD9567 and prednisolone.

Prednisolone dose (mg)	Average inhibition of TNFα over 24 h	Equipotent dose of AZD9567 (mg) Median (CI)
5	17 %	12 (8 – 19)
20	43 %	40 (29 – 54)
40	56 %	72 (52 – 99)
60	64 %	108 (76 – >155)

Prednisolone doses, estimated resulting TNFα inhibition, and estimated equipotent doses of AZD9567.

- Dose-response for AZD9567 and prednisolone was simulated using the PKPD model (upper left).
- Equipotent doses were identified based on equal levels of TNFα inhibition (lower left). For example, it was estimated that 40 mg AZD9567 (95% CI 29 – 54 mg) is equipotent to 20 mg prednisolone, with both doses resulting in an average TNFα inhibition of 43%.
- When performed over the whole range of studied doses, such comparisons defined the full equipotency relationship (lower right).

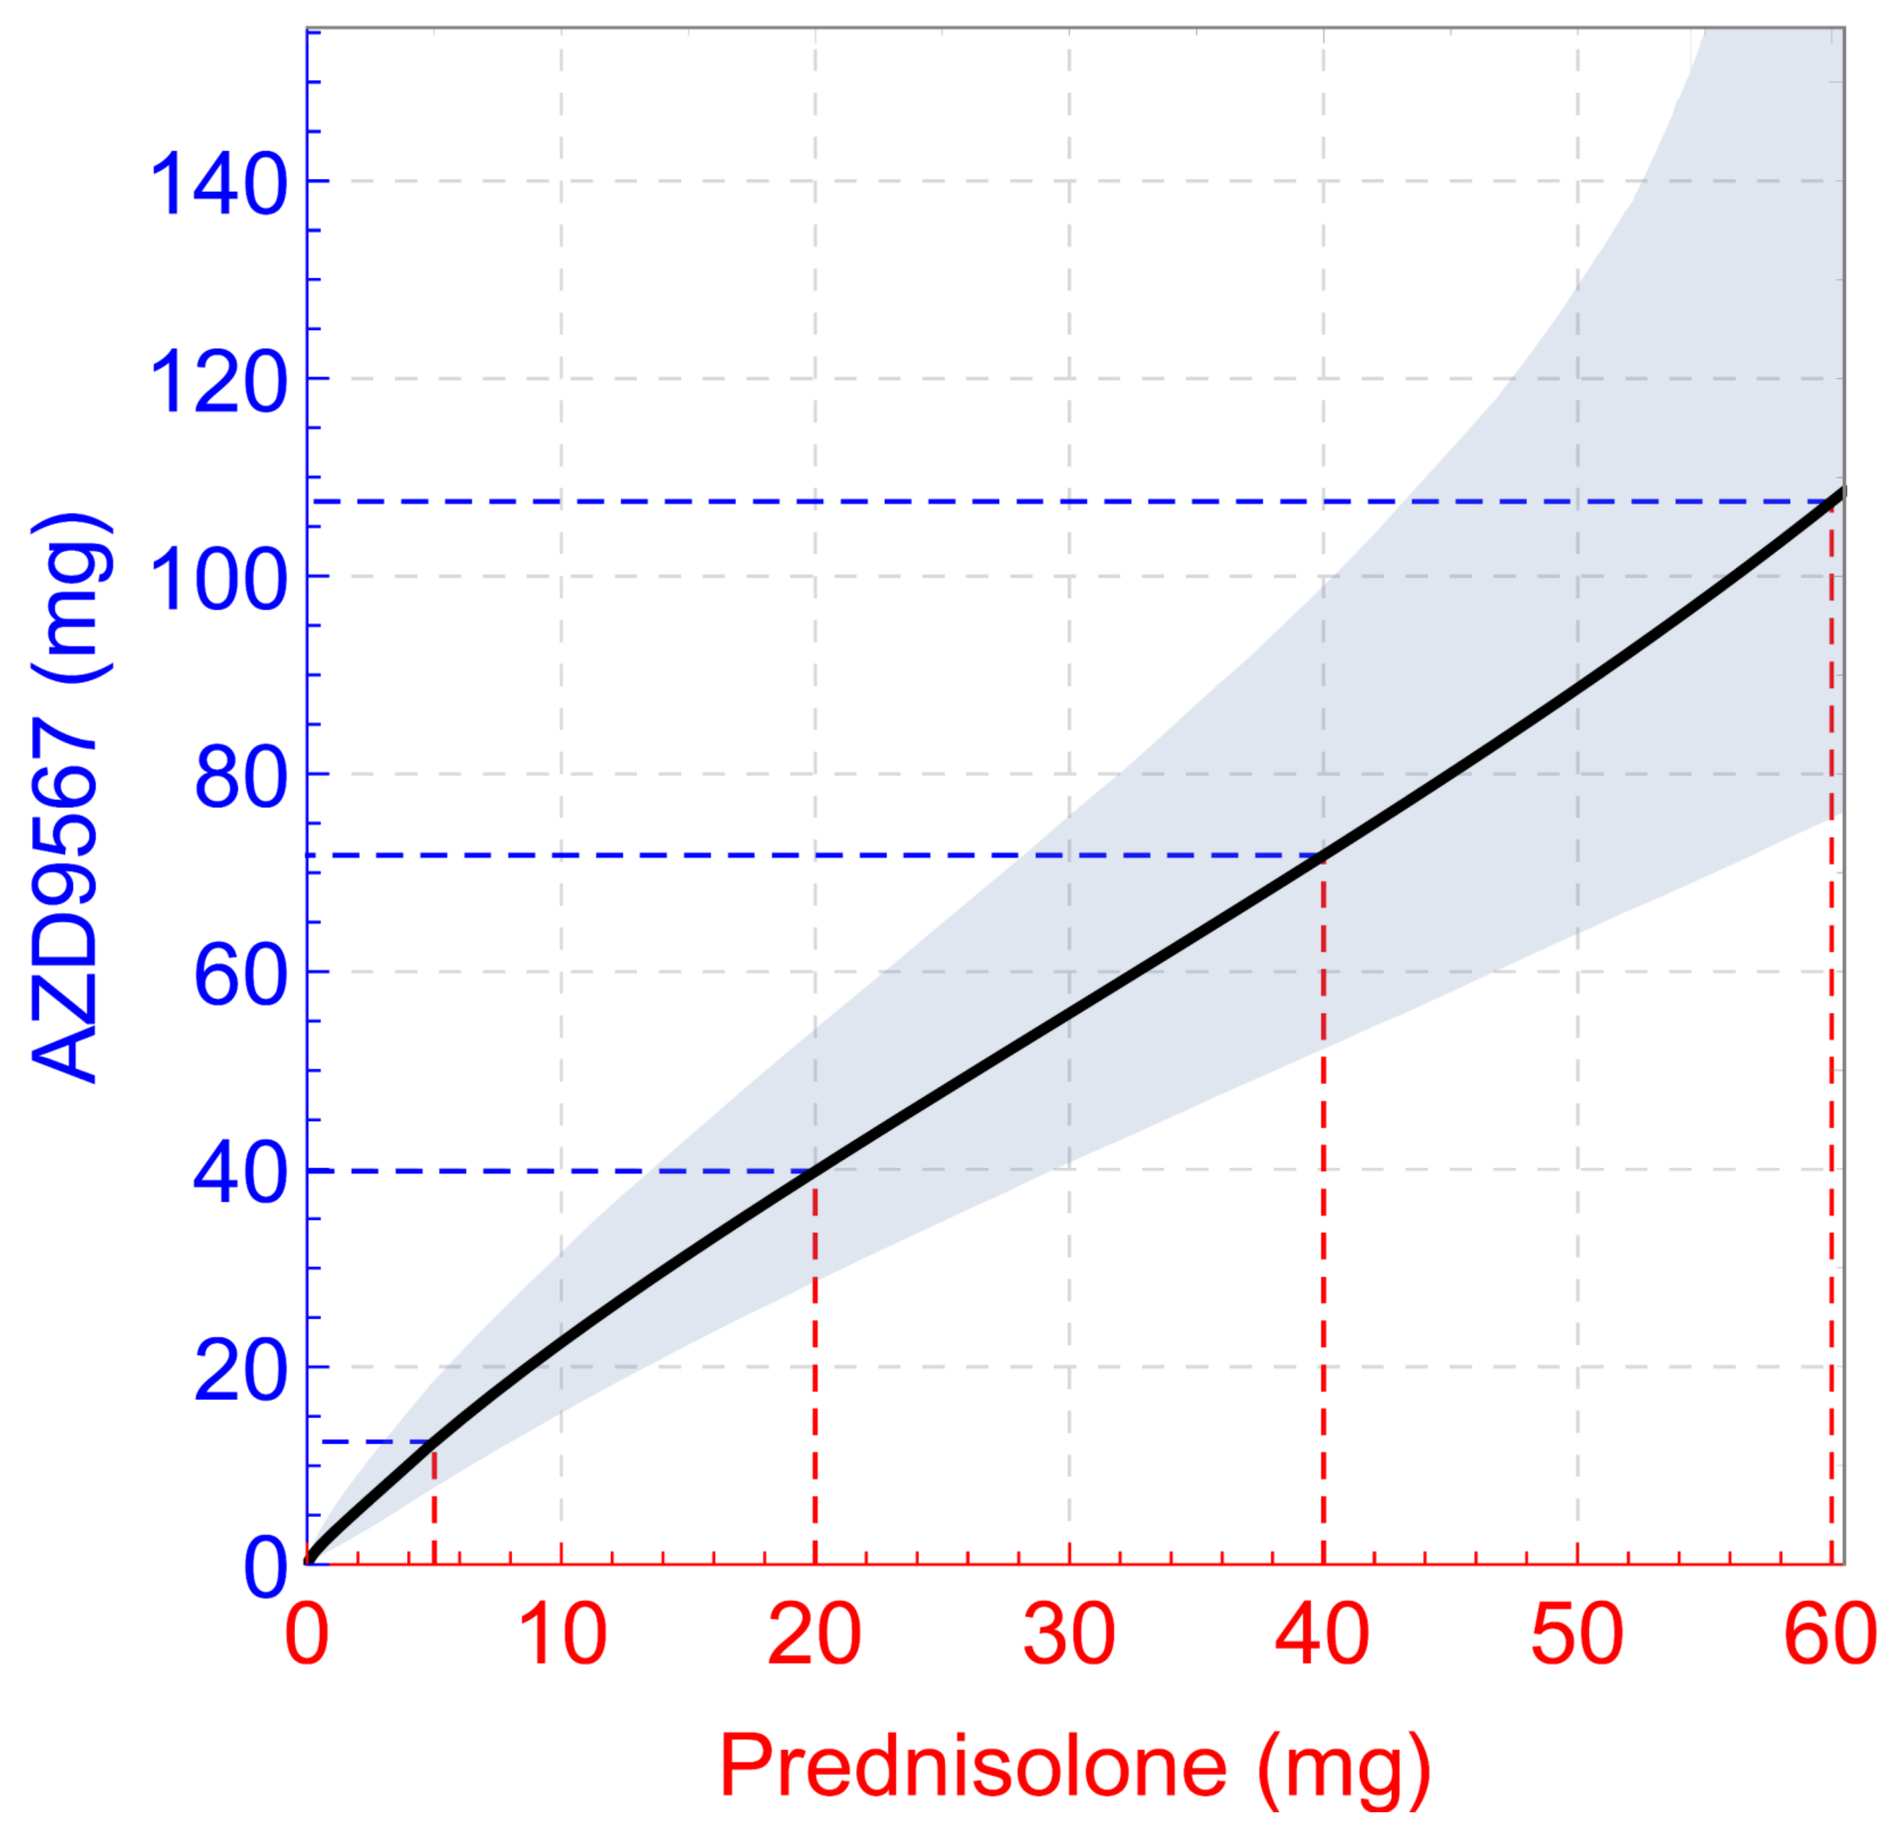
AZD9567 (mg)
Prednisolone (mg)

Estimated equipotency relationship of AZD9567 and prednisolone, with doses from the table highlighted.

Models

PK model for AZD9567

- A two-compartment model with first order absorption and a lag time was used to describe the PK of AZD9567.
- The dominant phase had a half-life of 4.4 h.
- Since the PK was not dose proportional, the oral dose was used in a linear covariate model for the relative bioavailability.

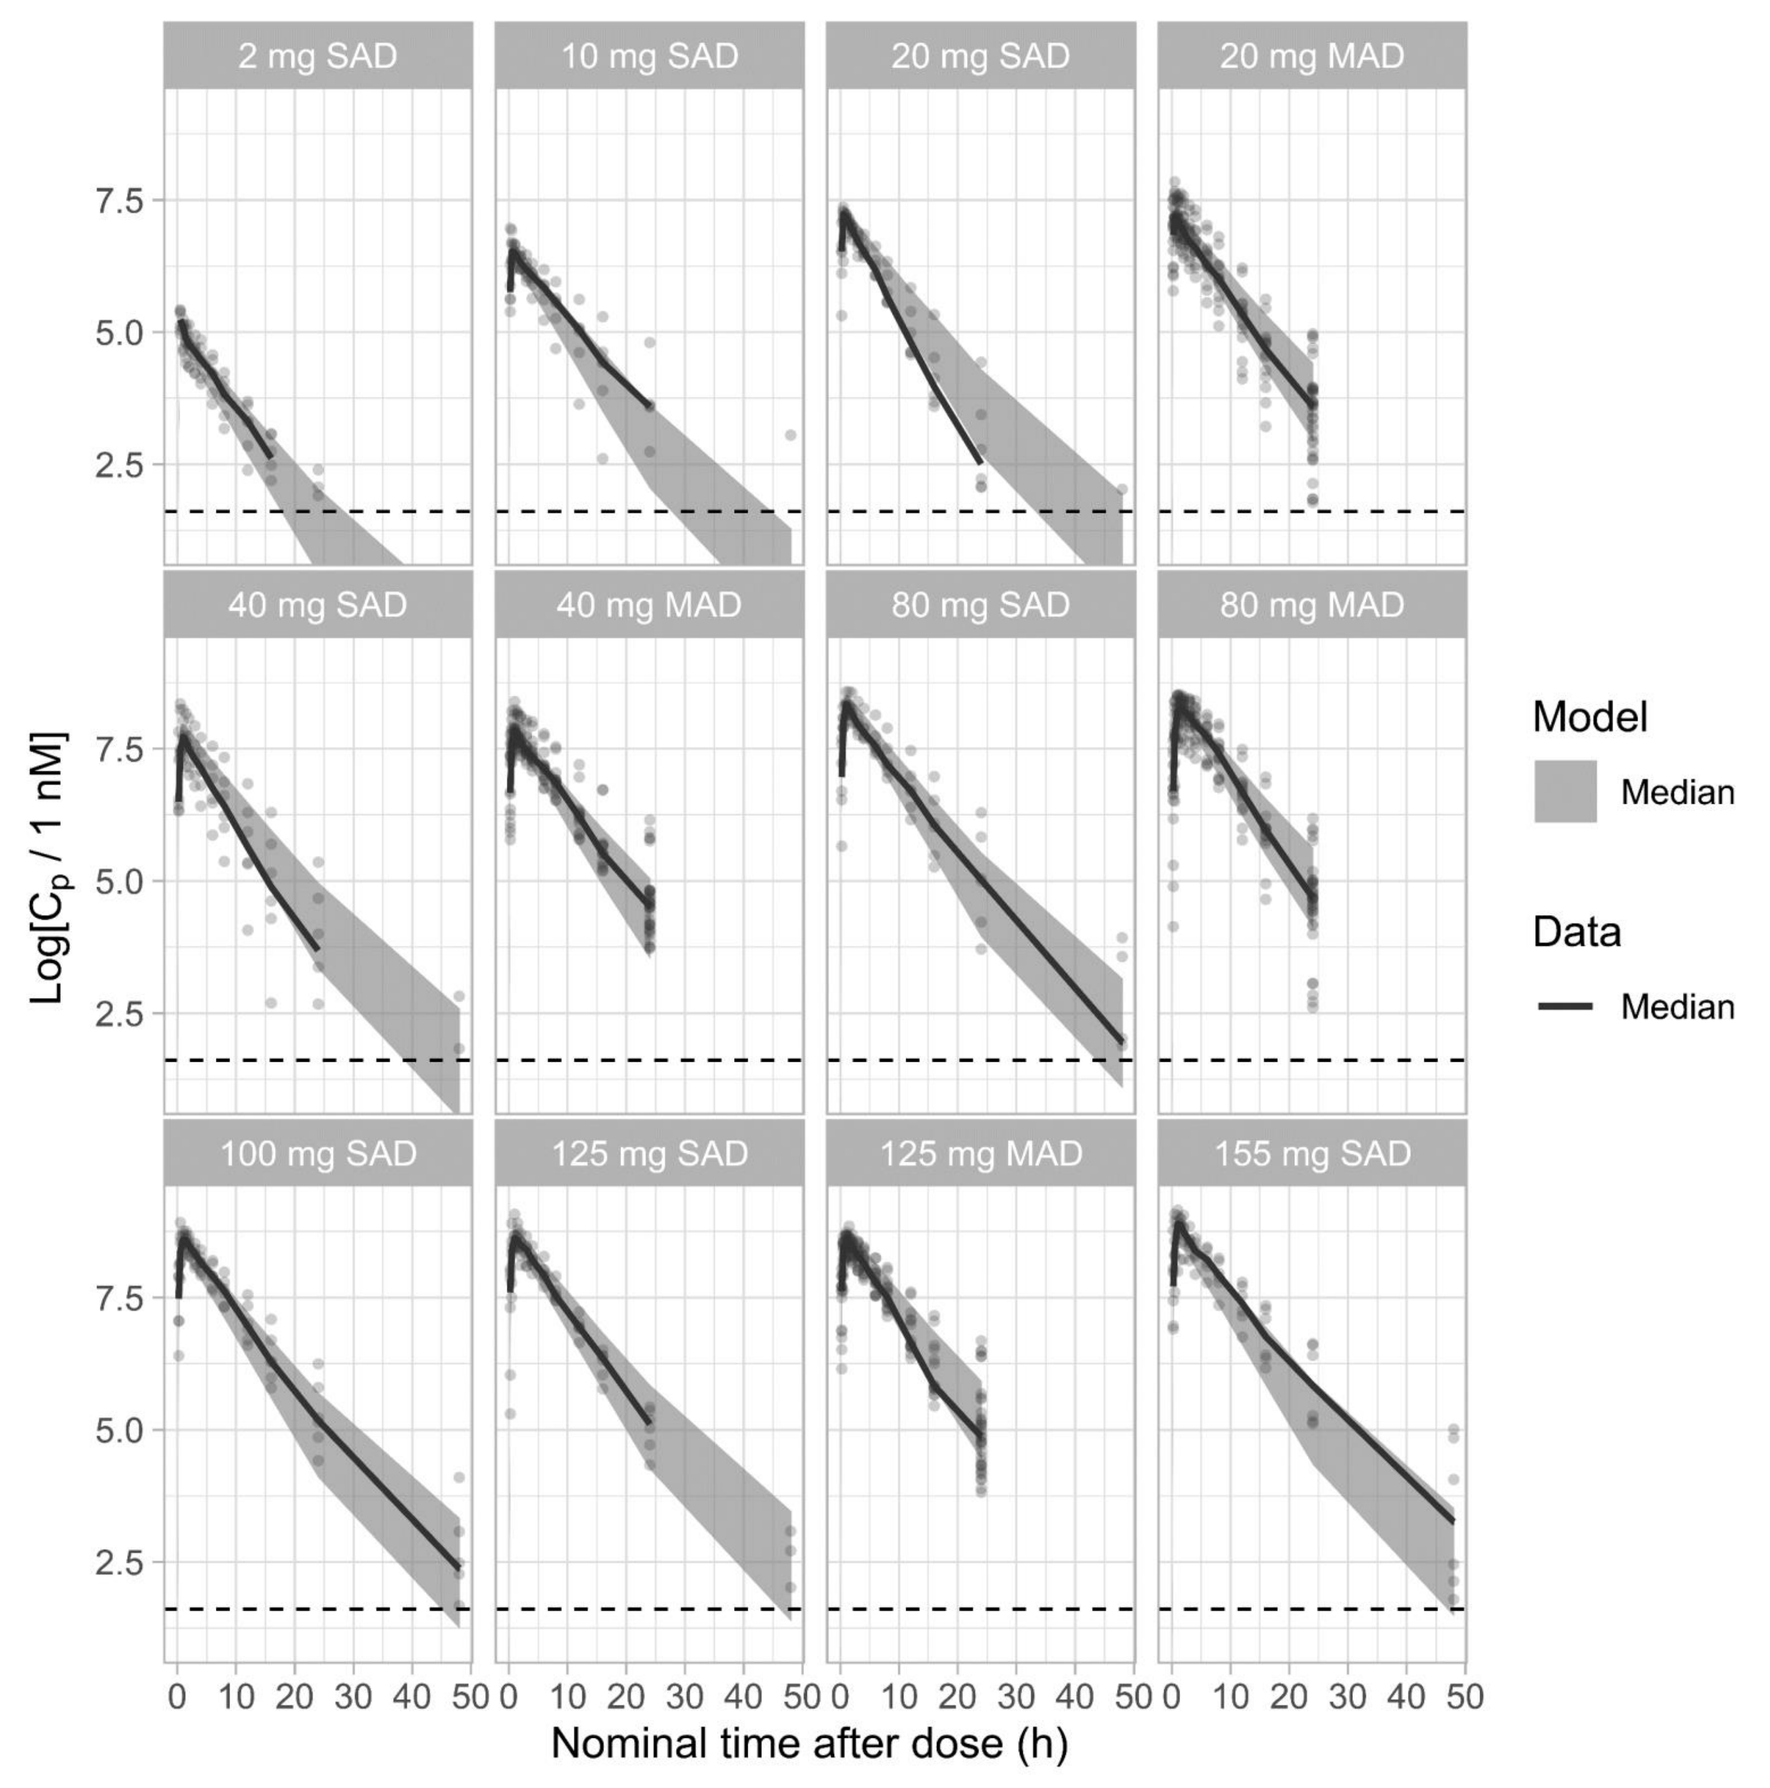
2 mg SAD
10 mg SAD
20 mg SAD
20 mg MAD
40 mg SAD
40 mg MAD
80 mg SAD
80 mg MAD
100 mg SAD
125 mg SAD
125 mg MAD
155 mg SAD
Log[Cp / 1 nM]
Nominal time after dose (h)
Model
Median
Data
Median

Treatment-stratified VPC for the AZD9567 PK model.

PK model for prednisolone

- Due to nonlinear plasma protein binding of prednisolone, total concentrations were converted into unbound concentrations using a static model.
- The unbound fraction goes from 6% in the limit of low concentrations to 38% in the high limit.
- A two-compartment model with a transit compartment approximation of the absorption [2] was used to model unbound prednisolone PK.
- The dominant phase had a half-life of 1.9 h.

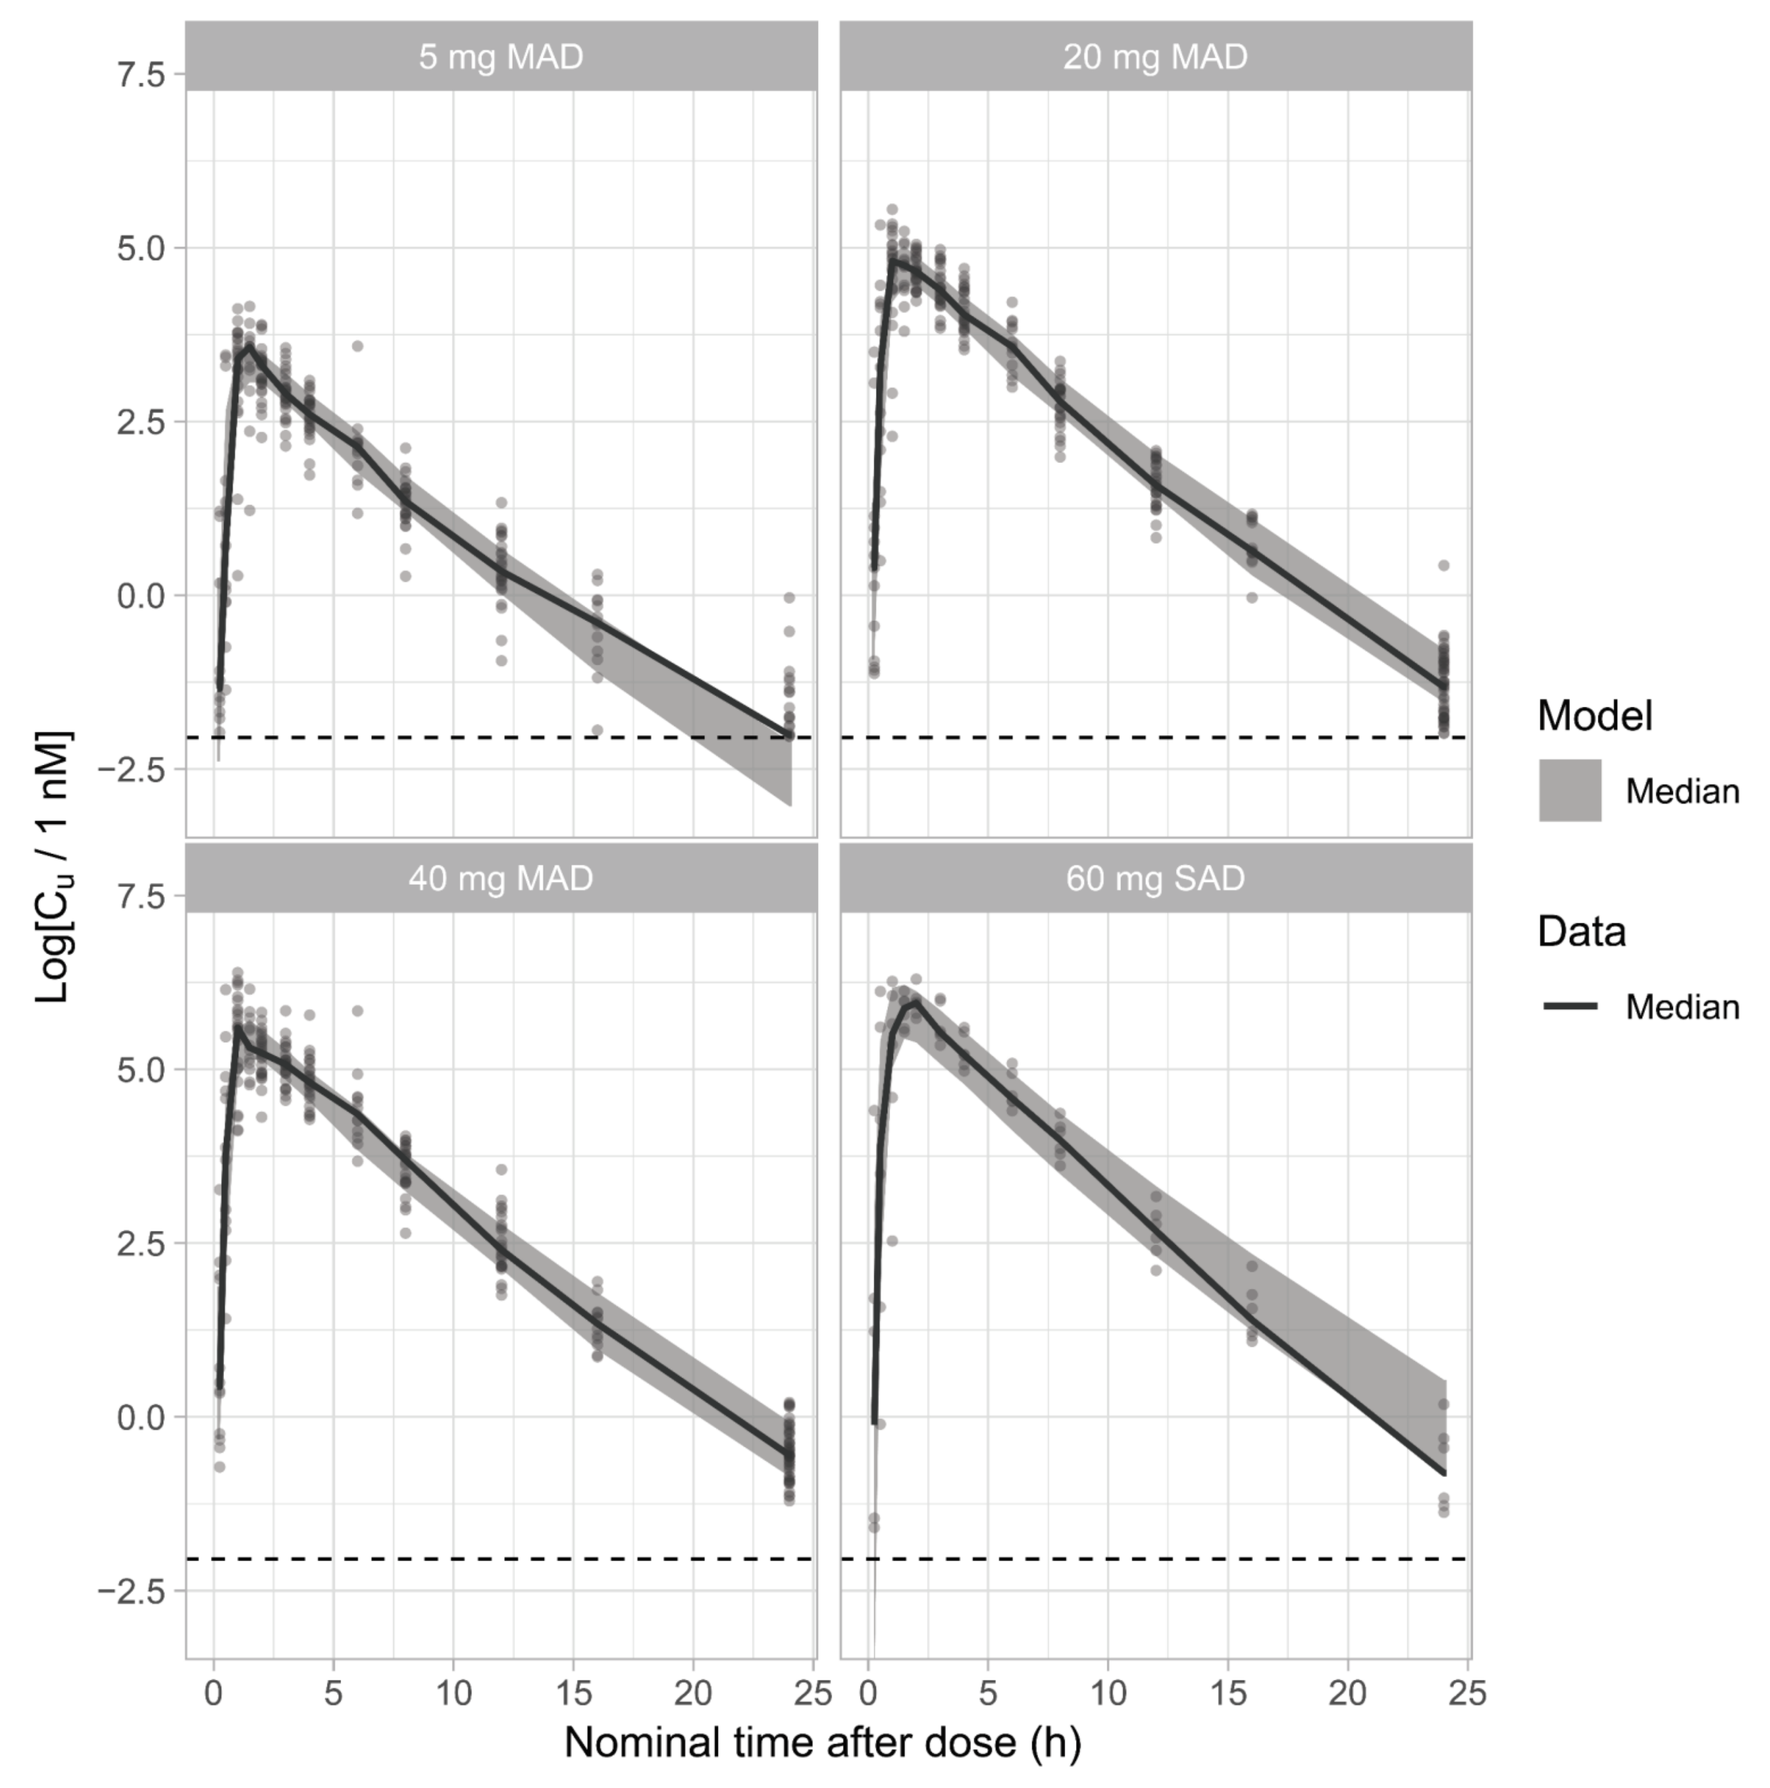
5 mg MAD
20 mg MAD
40 mg MAD
60 mg SAD
Log[Cu / 1 nM]
Nominal time after dose (h)
Model
Median
Data
Median

Treatment-stratified VPC for the prednisolone PK model.

PD model for TNFα

- TNFα inhibition was modeled by a turnover model with an inhibitory Imax-model for each compound.
- IC50 for total AZD9567 was estimated to 765 nM (95% CI of 610 – 920 nM). Given an unbound fraction 0.637%, this corresponds to a IC50 of 4.87 nM for unbound AZD9567.
- IC50 for unbound prednisolone was estimated to 17.0 nM (95% CI of 13.5 – 20.4 nM).

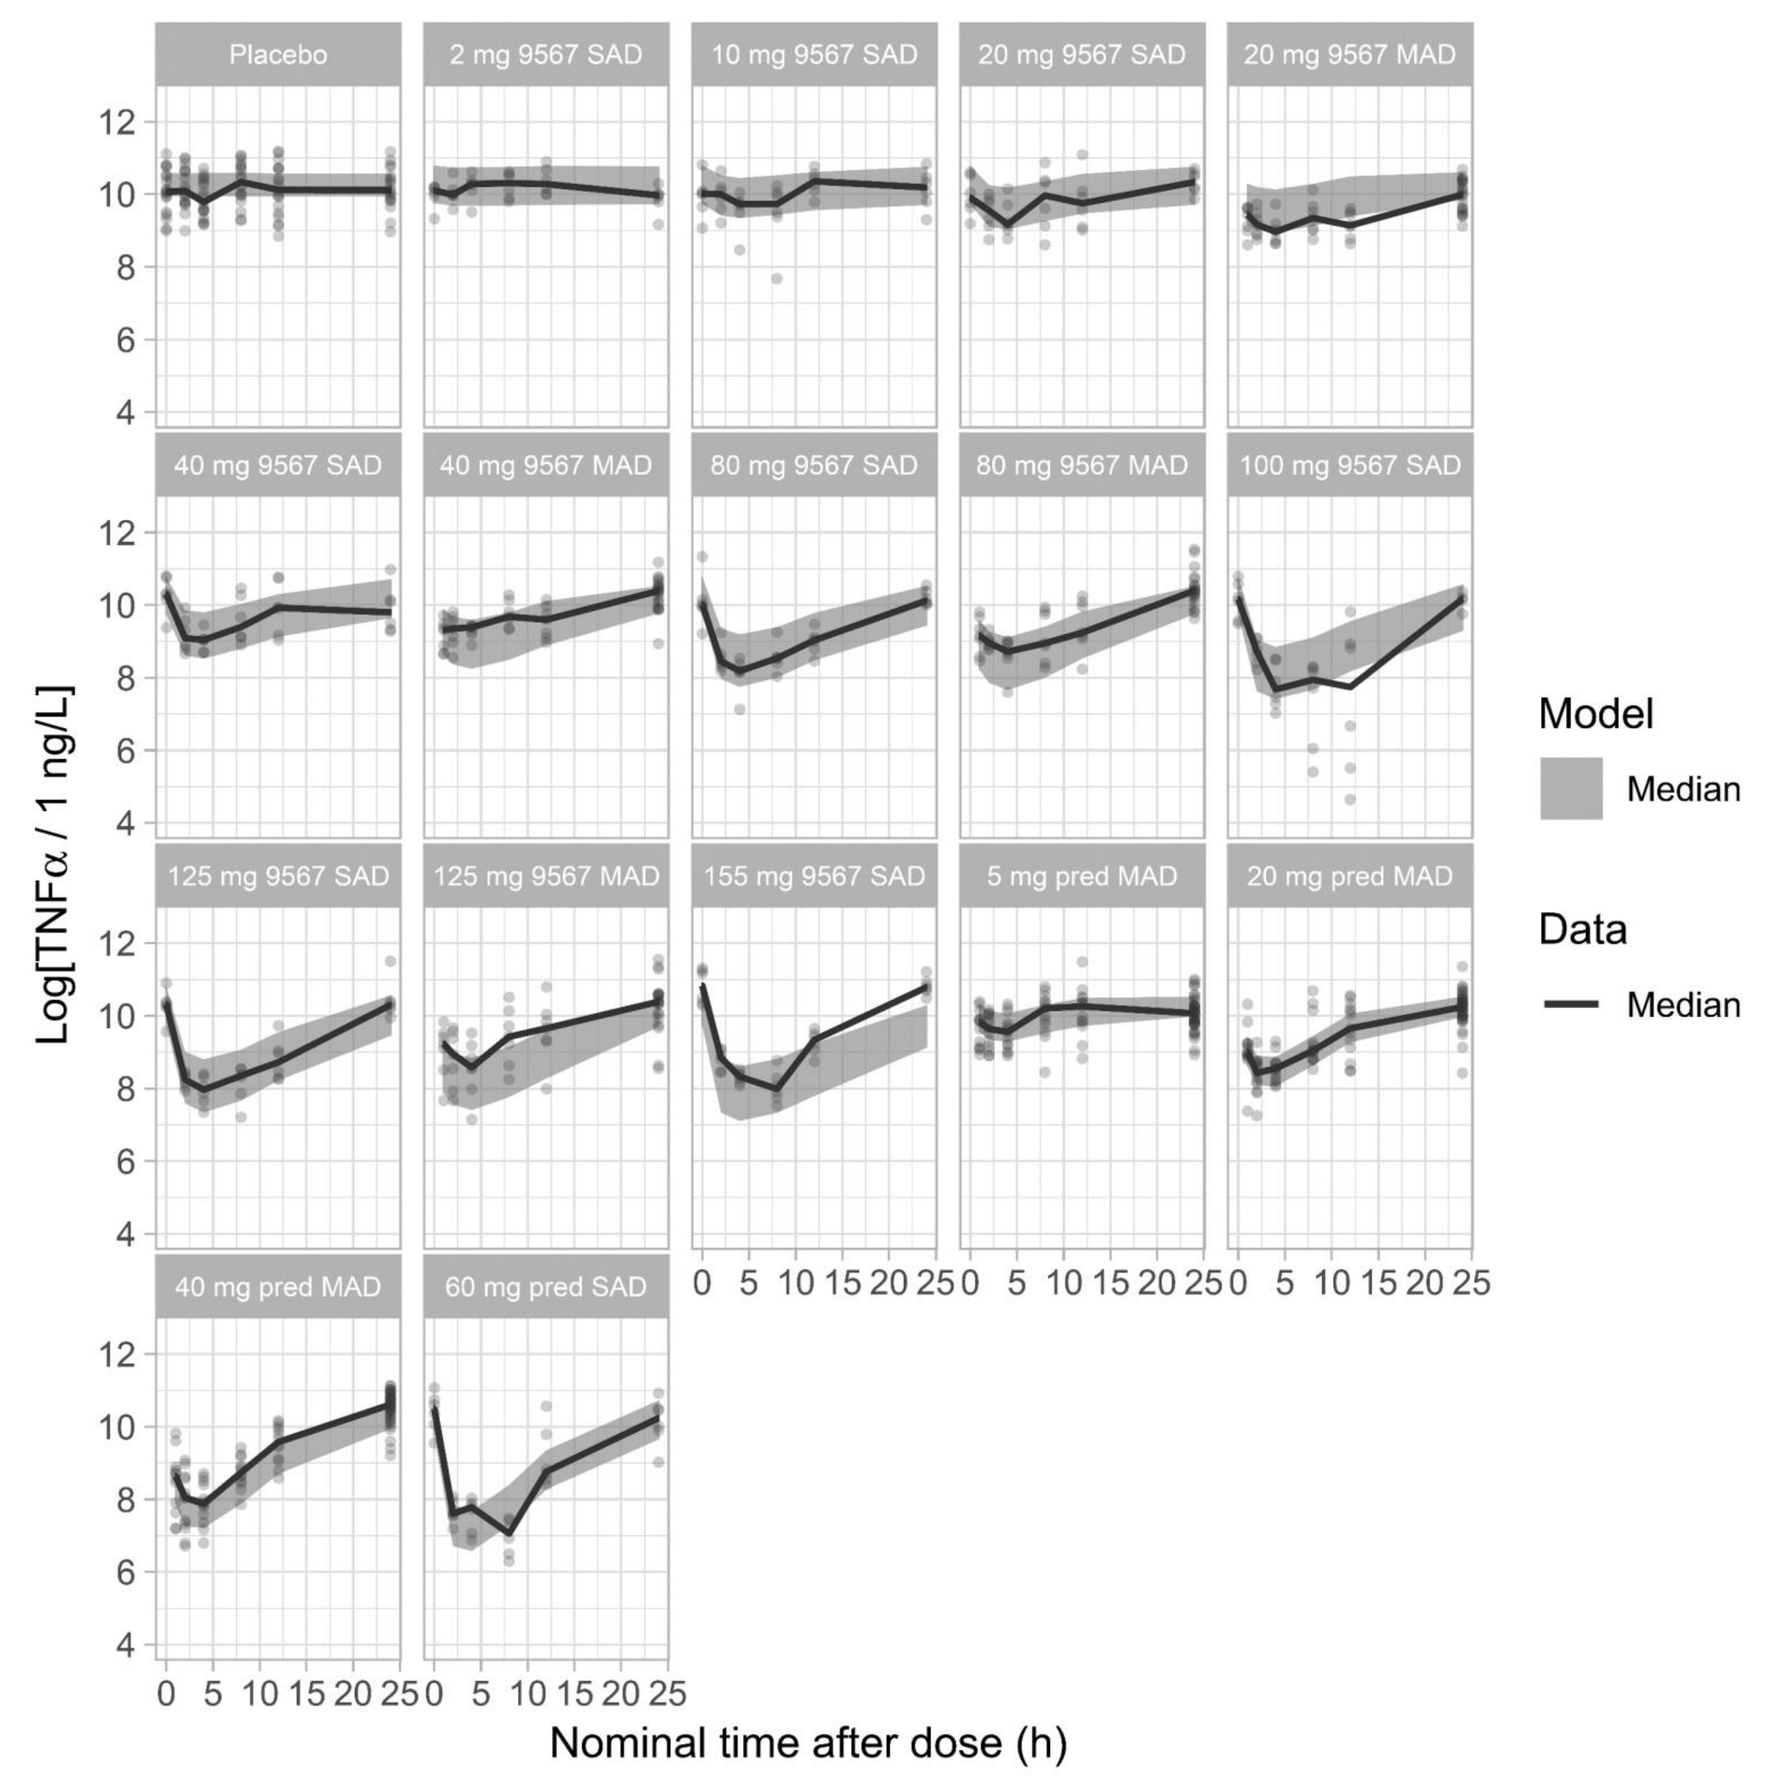
Placebo
2 mg 9567 SAD
10 mg 9567 SAD
20 mg 9567 SAD
20 mg 9567 MAD
40 mg 9567 SAD
40 mg 9567 MAD
80 mg 9567 SAD
80 mg 9567 MAD
100 mg 9567 SAD
125 mg 9567 SAD
125 mg 9567 MAD
155 mg 9567 SAD
5 mg pred MAD
20 mg pred MAD
40 mg pred MAD
60 mg pred SAD
Log[TNFα / 1 ng/L]
Nominal time after dose (h)
Model
Median
Data
Median

Treatment stratified VPC for the TNFα PD model.

Methods

- Models were based on clinical data from a single ascending dose (SAD) study [3] and a multiple ascending dose (MAD) study [4], measuring plasma concentration of the respective compound and concentration of the cytokine TNFα as released in response to an ex vivo whole blood lipopolysaccharide (LPS) stimulation [5].
- Nonlinear mixed effect PKPD modeling was done sequentially, fixing both fixed and random effects of the PK models before fitting the PD model.
- All models were estimated with NONMEM 7.3.0 using the FOCE method [6].
- The PD model contained both common and compound-specific parameters and was fitted for both compounds simultaneously.
- Dose-response curves for each compound were determined by simulation.
- The response was defined as the average inhibition of TNFα over 24 h following 5 consecutive daily doses.
- Confidence intervals for the equipotent dose relationship were determined by simultaneously accounting for the uncertainty in the dose-response of each compound.

References

1. Ripa et al. Discovery of a novel oral glucocorticoid receptor modulator (AZD9567) with improved side effect profile. J Med Chem 2018; 61(5):1785-1799
2. Savic R, Jonker D, Kerbusch T, Karlsson M. Implementation of a transit compartment model for describing drug absorption in pharmacokinetic studies. J Pharmacokinet Pharmacodyn 2007; 34:711-726
3. AstraZeneca. A single ascending dose study to assess the safety, tolerability, pharmacokinetics and pharmacodynamics of AZD9567. Available from: https://clinicaltrials.gov/ct2/show/NCT02512575
4. AstraZeneca. A study to assess the safety, tolerability, pharmacokinetics and pharmacodynamics of multiple ascending doses of AZD9567. Available from: https://clinicaltrials.gov/ct2/show/NCT02760316
5. Schmolz M and Eisinger D. TruCulture®: A simple whole blood collection and culture system for quantifying physiological interactions of the human immune system in the clinic.
6. Beal S and Sheiner L, "NONMEM User's Guides. (1989-2009)" Icon Development Solutions, Elliot City, MD, USA, 2009.

Supported by AstraZeneca

Presented at PAGE 2019, Stockholm Watergate Congress Centre, Stockholm, June, 2019